Partnerships for Global Vaccine Development Rotavirus Vaccine

Partnerships for Global Vaccine Development - Meningitis Vaccine Project (MVP) for Sub Saharan Africa

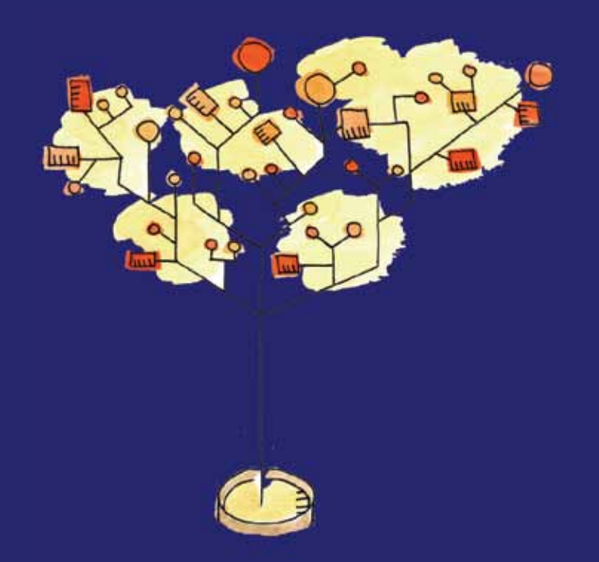

Kevin Chang, Ph.D.; Peter Soukas. J.D.; Susan Ano, Ph.D, Uri Reichman, Ph.D.

Office of Technology Transfer, National Institutes of Health, U.S. Department of Health and Human Services, CSTA Industry Forum, Bethesda, MD, February 17-18, 2010

Rotavirus

- Rotavirus is the most common cause of severe diarrhea among children
 - resulting in the 2,000,000 hospitalizations every year worldwide.
 - hospitalization of approximately 55,000 children each year in the United States.
 - deaths of over 500,000 children annually worldwide.
 - 80% of deaths of children occur in poor countries.

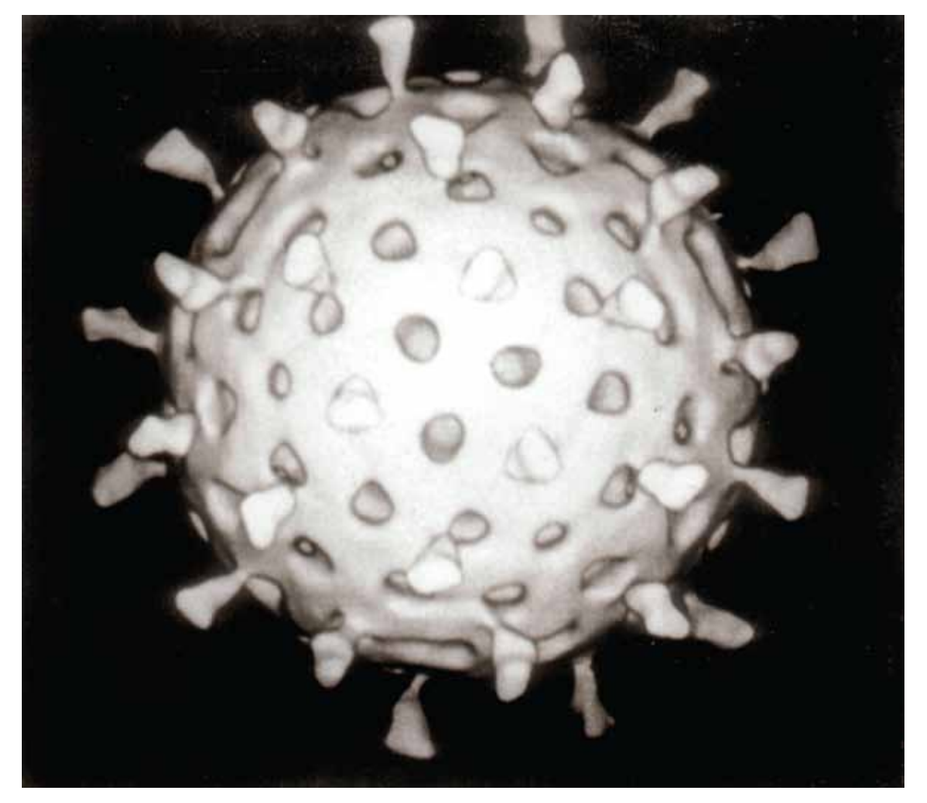

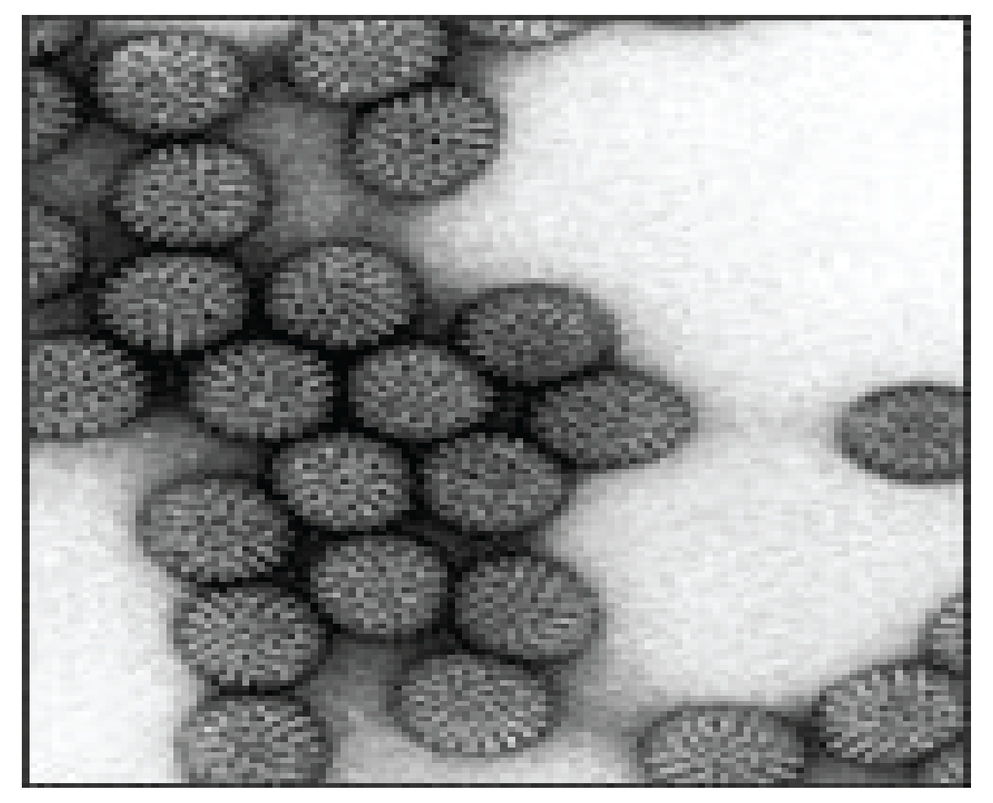

Rotavirus Vaccine: NIH Partnerships

From Sabin Vaccine Institute and WHO
- Annually, over 500,000 deaths worldwide

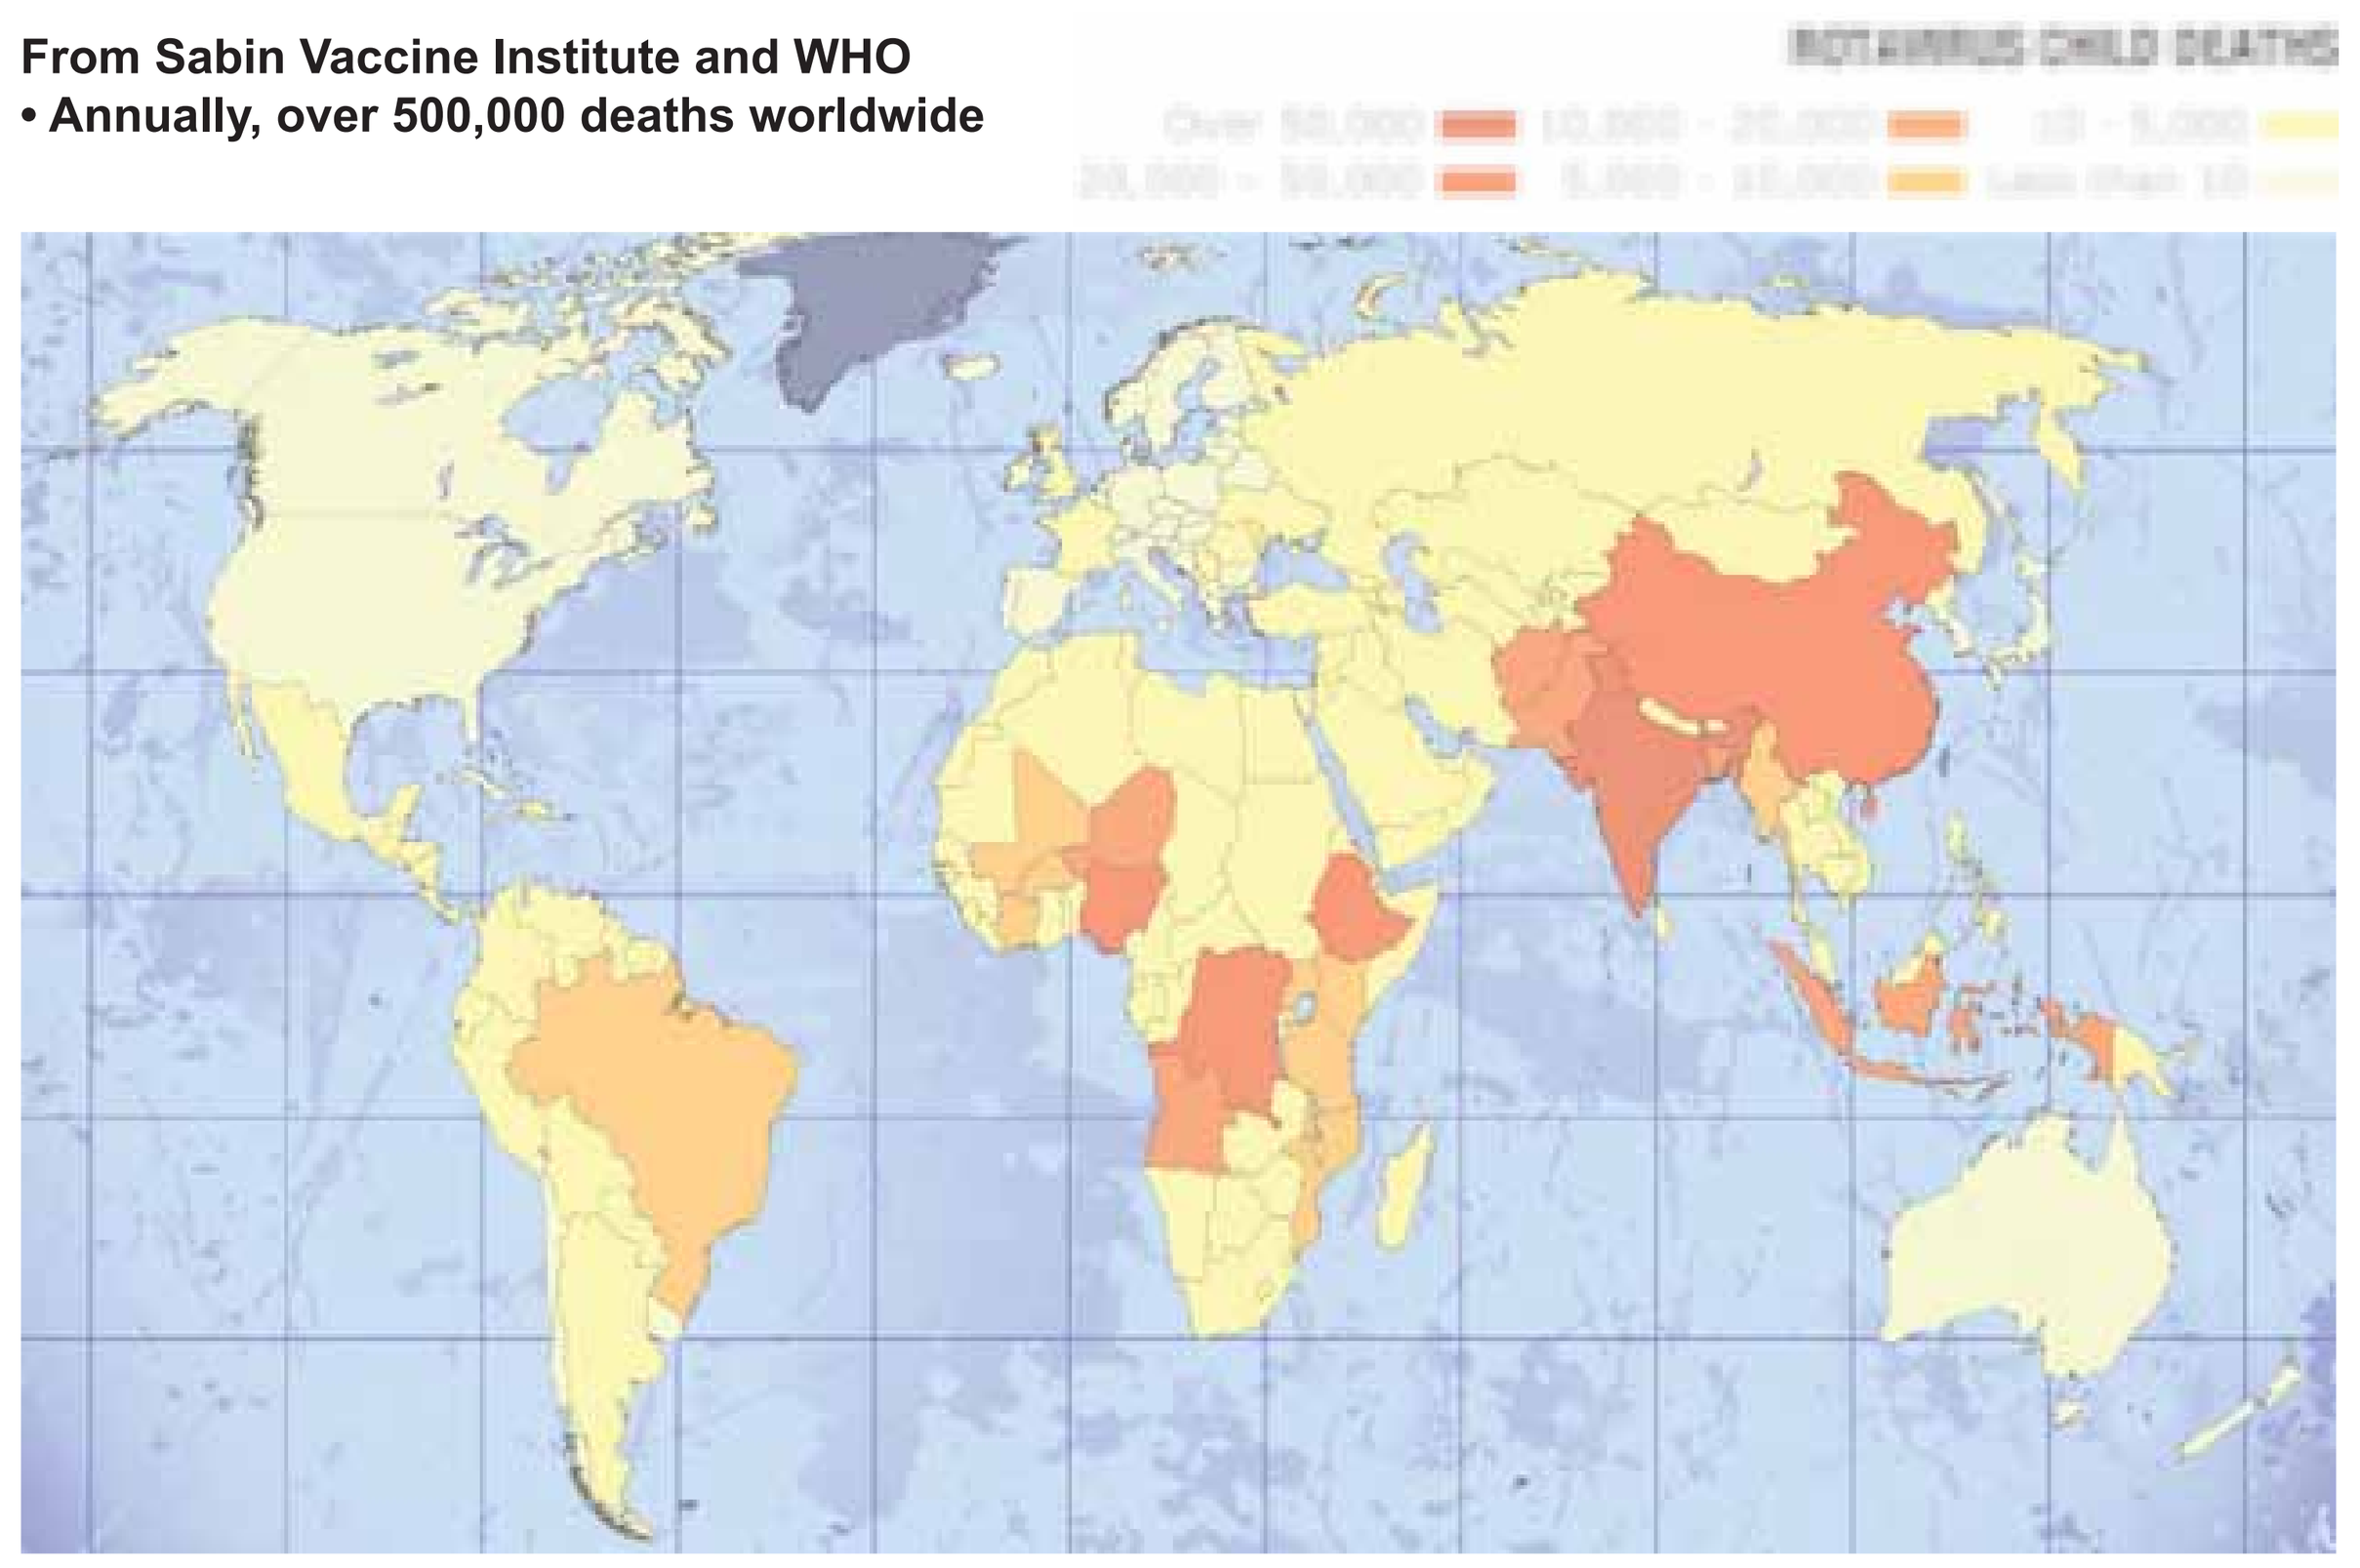

The Partnership for Global Development

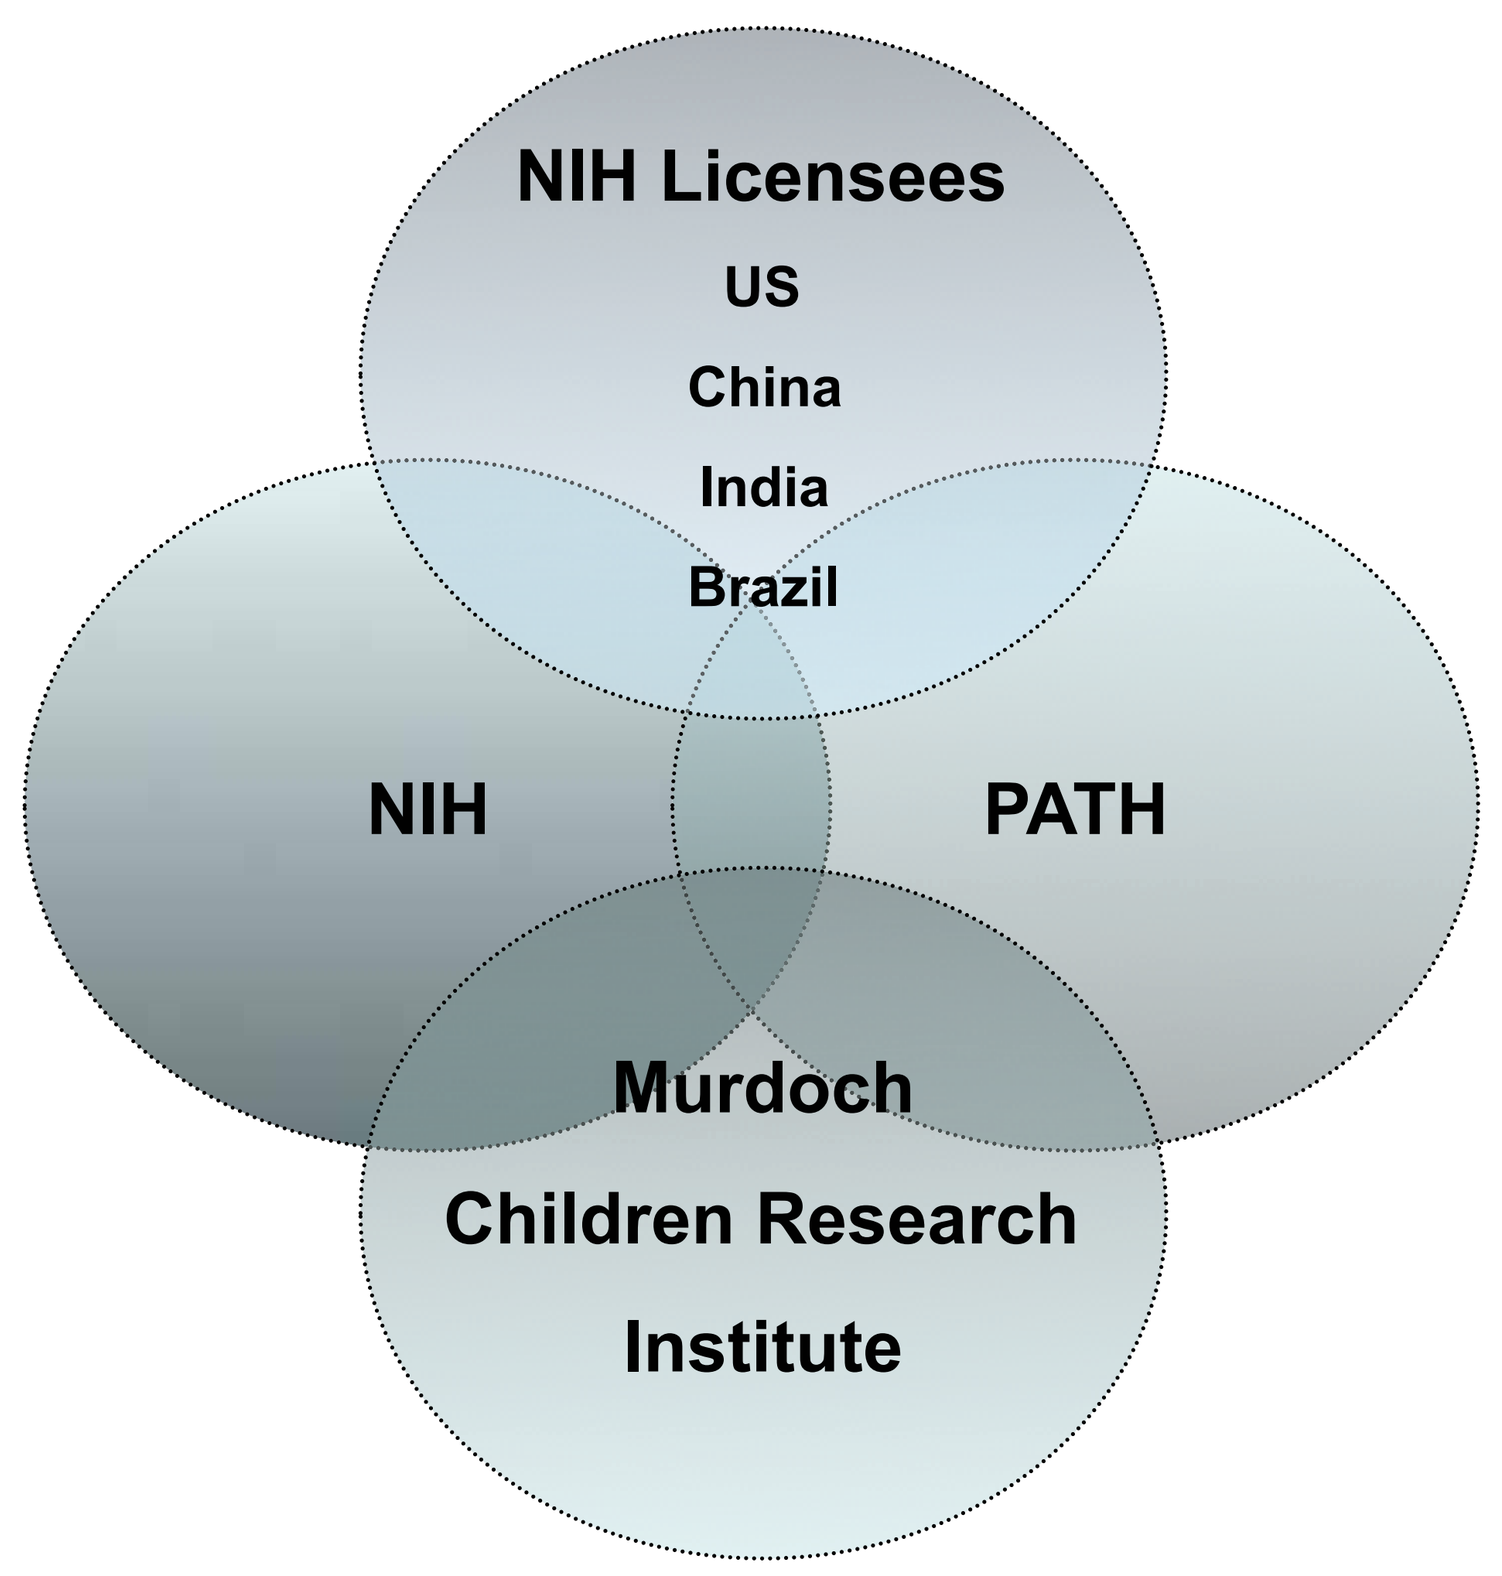

NIH Licensees
US
China
India
Brazil
NIH
PATH
Murdoch
Children Research
Institute

Role of Partnership Components

<u>NIH</u>
- Provided the IP/technology
- Provided biological materials
 - Virus strains
 - Antibodies
- Licensed to commercial organizations

<u>PATH</u>
- License Agreement with the NIH
- Funding support to NIH licensees
 - $19.1M; 2006-2011
 - For scale-up activities
 - For clinical trials

<u>NIH Licensees</u>
- China
- India
- Brazil
- USA

<u>Murdoch Children Research Institute (Australia)</u>
- Scale-up agreement with PATH
- Scale-up production of virus strains and production of antibodies
- Provide scale-up materials to NIH licensees for clinical studies

NIH Licensees for Rotavirus Vaccine Technology

- **India**
 - Shantha Biotechnics Pvt. Ltd.
 - Bharat Biologicals International Ltd.
 - Biological E Ltd.
 - Serum Institute of India Ltd. (SII)
- **China**
 - Wuhan Institute for Biological Products
 - Sinovac Biotech Ltd.
 - Xinkexian (Beijing) Biological Technology Co., Ltd.

Brazil and Latin America
- Fundação Butantan

North American and Europe - Aridis Pharmaceuticals

Regional Exclusive licenses. Non-exclusive for rest of the world

Rotavirus Vaccine Project Technology Background

- Hexavalent Human-bovine reassortants viruses: G1-G4; G8, G9
- IP rights in US, Europe, Japan, Canada, Australia, Brazil, China, India, and South Korea

- NIH inventor: Dr. Albert Kapikian
 - Expert in the field
 - Provides continuous sdvise to licensees

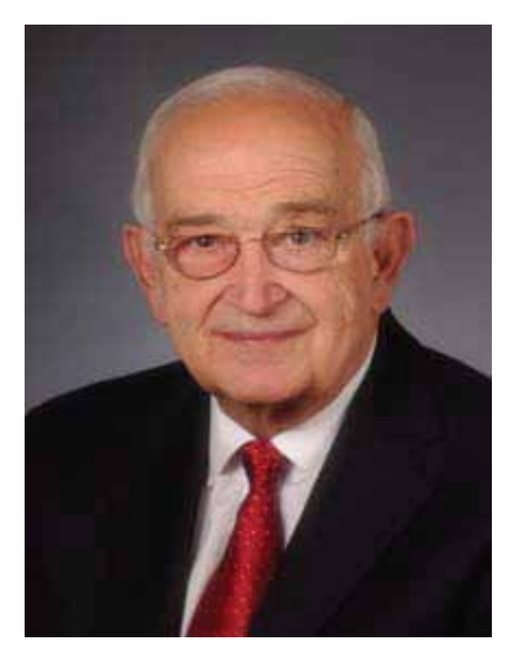

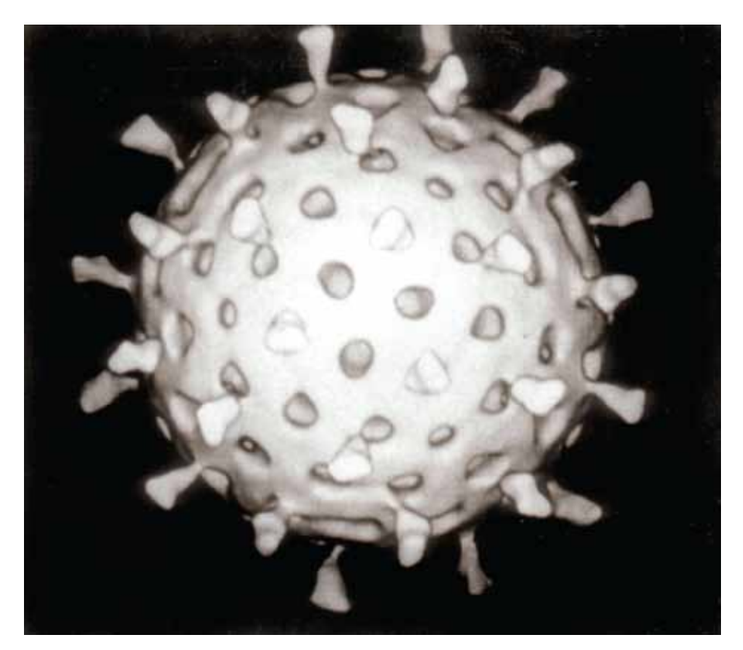

PATH – Program for Alternative Technology in Health A Catalyst for Global Health

- Global Charitable Organization
 - Focus on global health
 - A component of Bill and Melinda Gates Foundation

- Creates Global Partnerships
 - Vaccine development for developing countries
 - Financial support of the global partnership

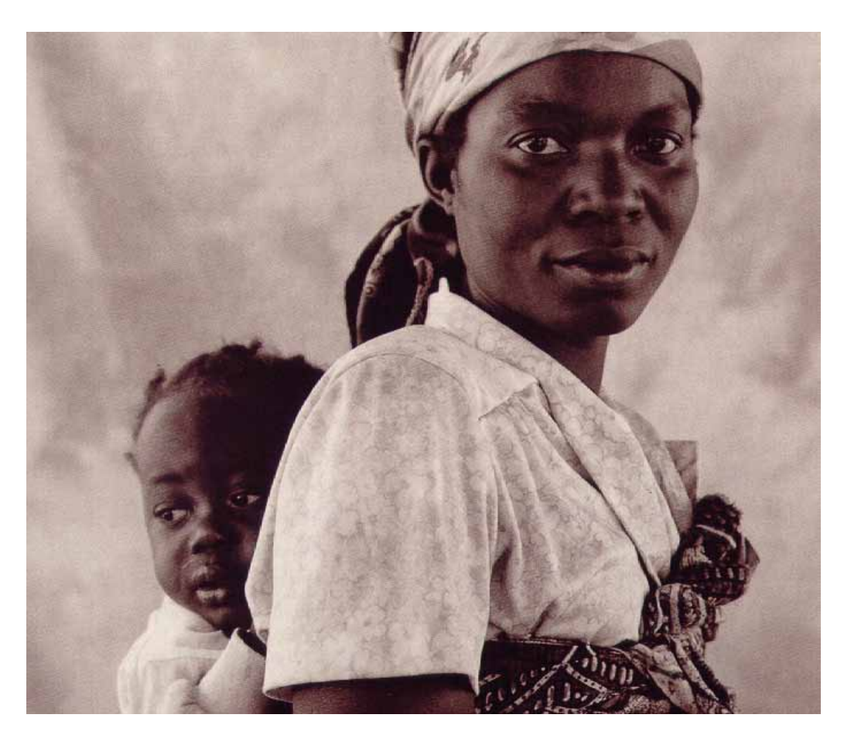

Principles of the Rotavirus Partnership

- Continuous interactions between all the parties
- Continuous report of progress by licensees within the obligations of the license agreements
- Annual meetings with the participation of all parties including consultants
 - Share information
 - Reporting progress
 - Discuss and resolve technical issues
 - Decide on future steps
- NIH inventor continued involvement with licensees through consultation and advice

Meningitis Vaccine Project

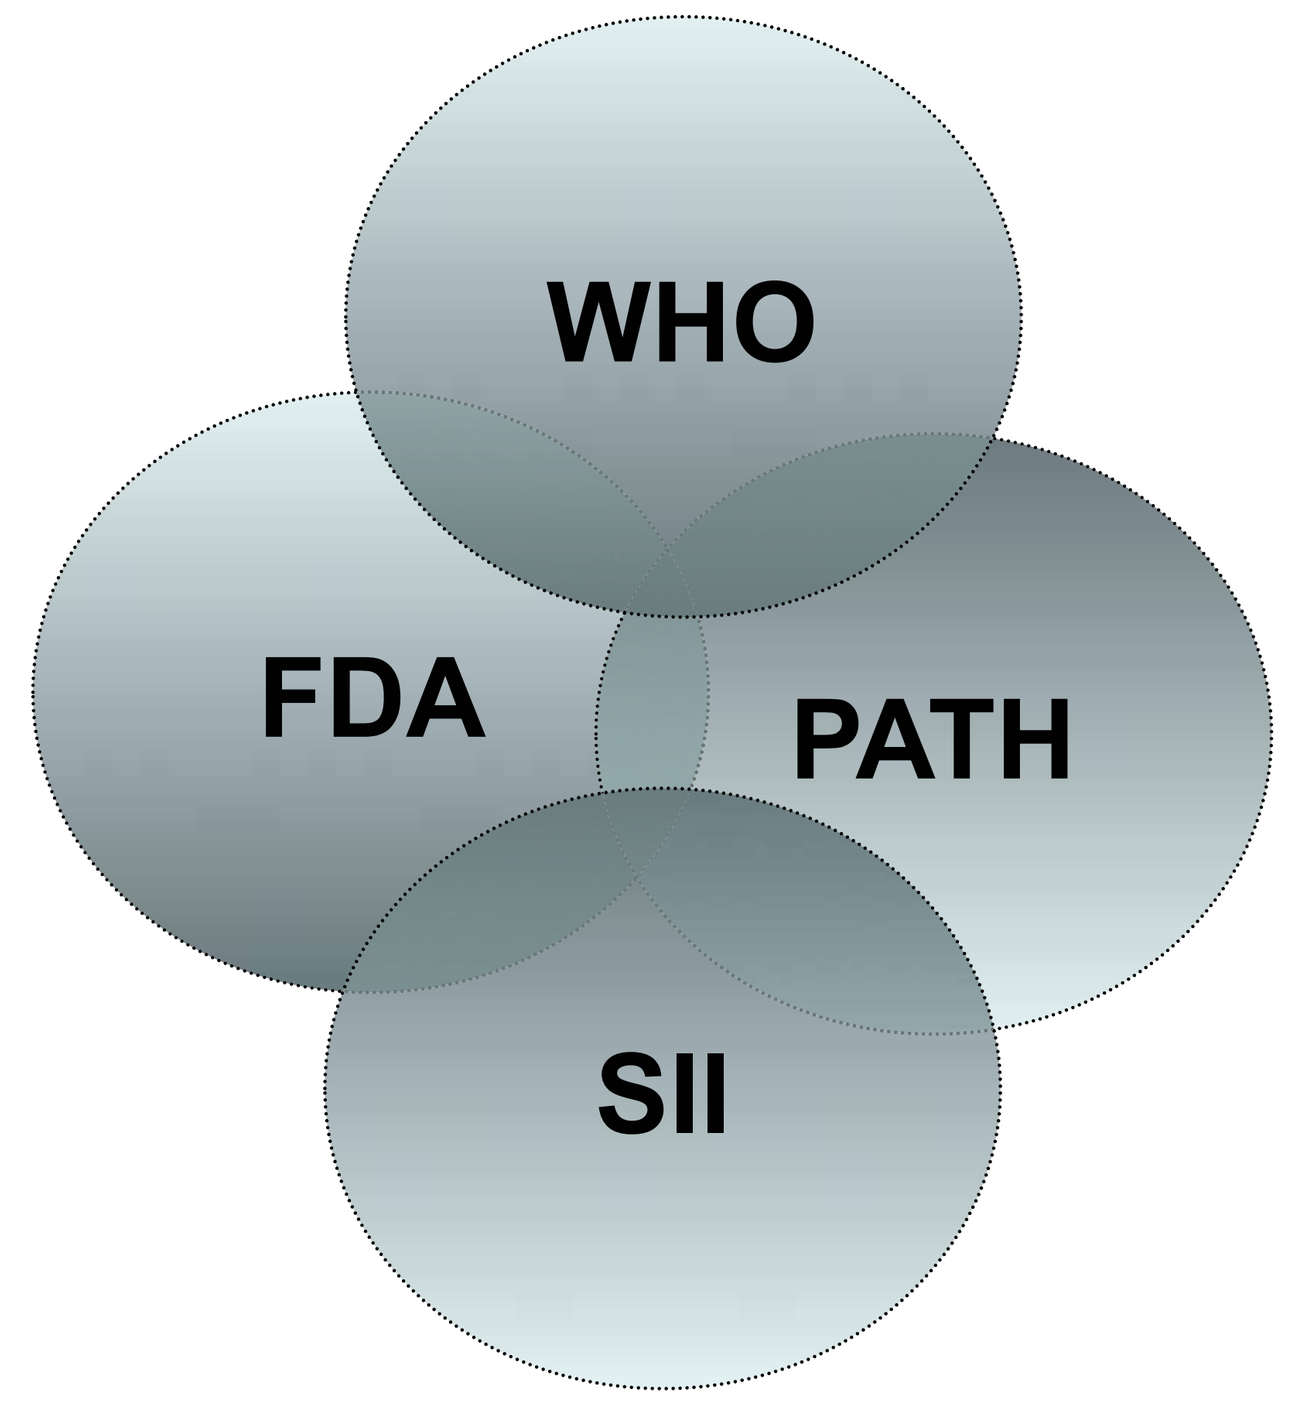

WHO
FDA
PATH
SII

Meningitis Vaccine for Sub Saharan Nations: Partnership between WHO and PATH with IP from FDA

Bill & Melinda Gates Foundation

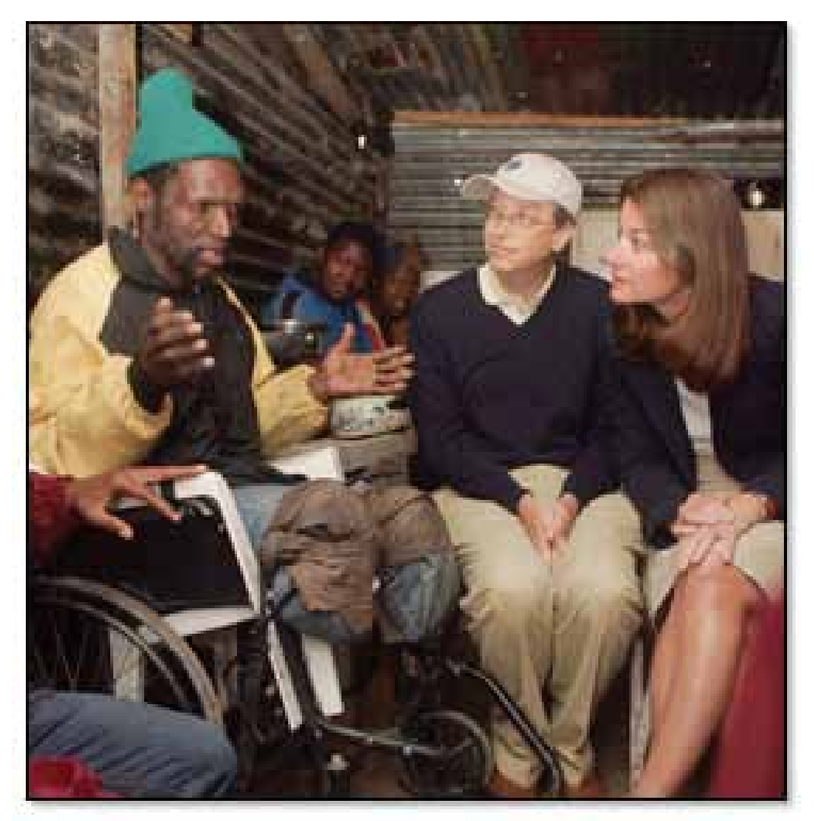

Meningitis Vaccine Projcet (MVP) PATH

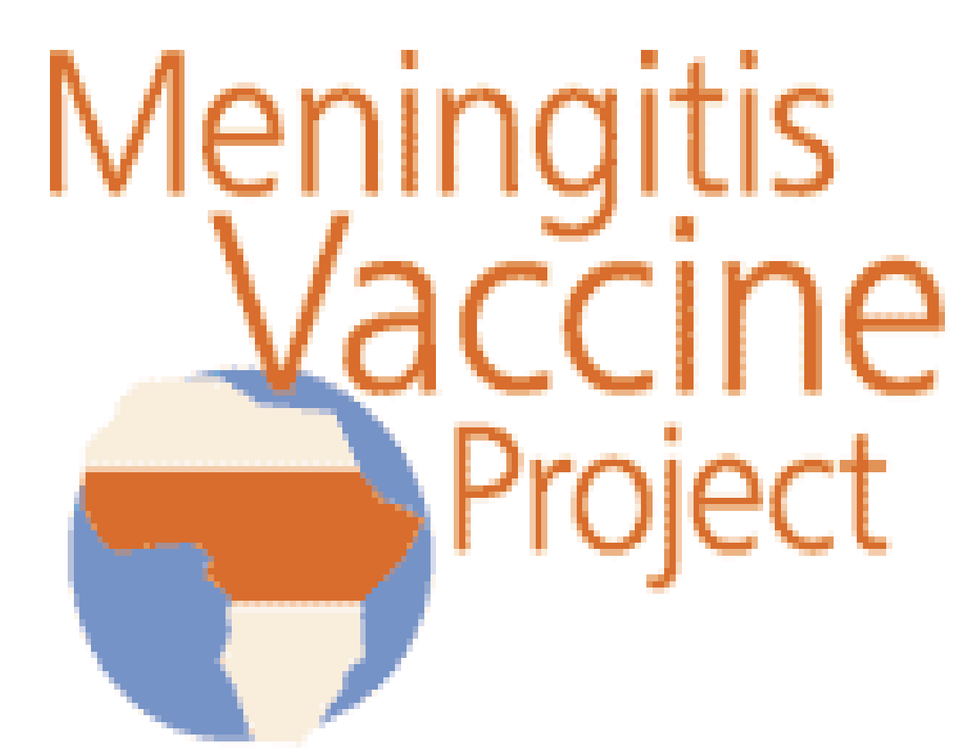

Meningitis
Vaccine
Project

Meningitis

- Meningitis is an infection of the fluid of a person's spinal cord and the fluid that surrounds the brain
- Bacterial Meningitis can have devastating effects resulting in death or permanent brain and hearing damage
- Neisseria Meningiditis is the leading cause of Meningococcal disease
- Worldwide, the vast majority of disease is caused by 5 serogroups (A, B, C, Y, W-135) of the bacterium

Meningitis Belt in Africa

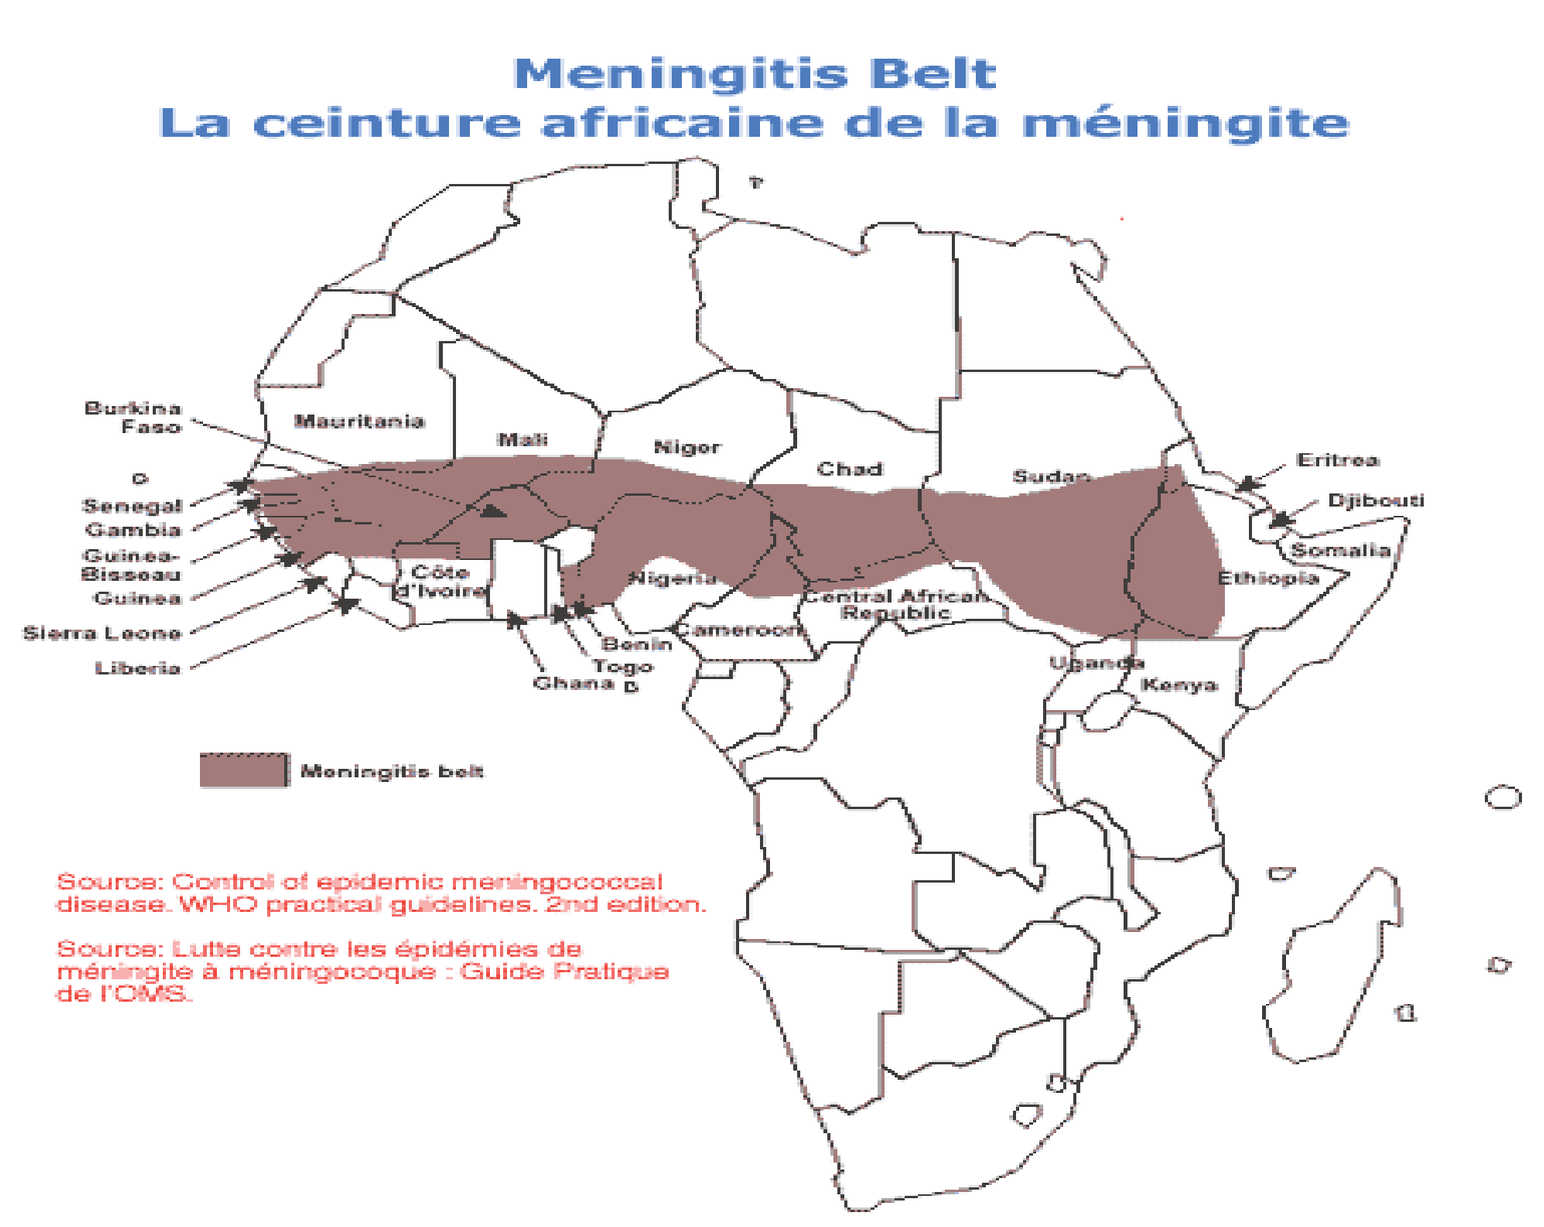

Meningitis Belt
La ceinture africaine de la méningite
Burkina Faso
Mauritania
Mali
Niger
Chad
Sudan
Eritrea
Djibouti
Somalia
Ethiopia
Senegal
Gambia
Guinea-Bisseau
Guinea
Sierra Leone
Liberia
Côte d'Ivoire
Nigeria
Benin
Togo
Ghana
Cameroon
Central African Republic
Uganda
Kenya
Meningitis belt
Source: Control of epidemic meningococcal disease. WHO practical guidelines. 2nd edition.
Source: Lutte contre les épidémies de méningite à méningocoque : Guide Pratique de l'OMS.

Meningitis Epidemic in Africa

- In the so called 'Menigitis belt' that stretches from Ethiopia to Gambia and Senegal, Meningococcal disease has killed more than 100,000 people between 1988 and 1997, with more than 704,000 cases reported to the World Health Organization.
- The disease develops at extremely high speed and the severity of the after effects can be harsh.
- At least 10 percent of infected individuals die during the acute episode.
- Some 10-20 percent of survivors develop permanent pathological damage such as epilepsy, hearing loss, or mental retardation.

- Existing vaccines based on bacterial polysaccharides are only partially effective due to three shortfalls:
 - They do not produce long-lasting protection
 - They do not protect very young children
 - They do not decrease transmission of the organism in the population.

Partnership Components

- Under the terms of the licensee agreement NIH-PATH, SII of India was granted sublicensing rights to scale up the procedure and to manufacture the vaccine
- The project is now in Phase III clinical trials.

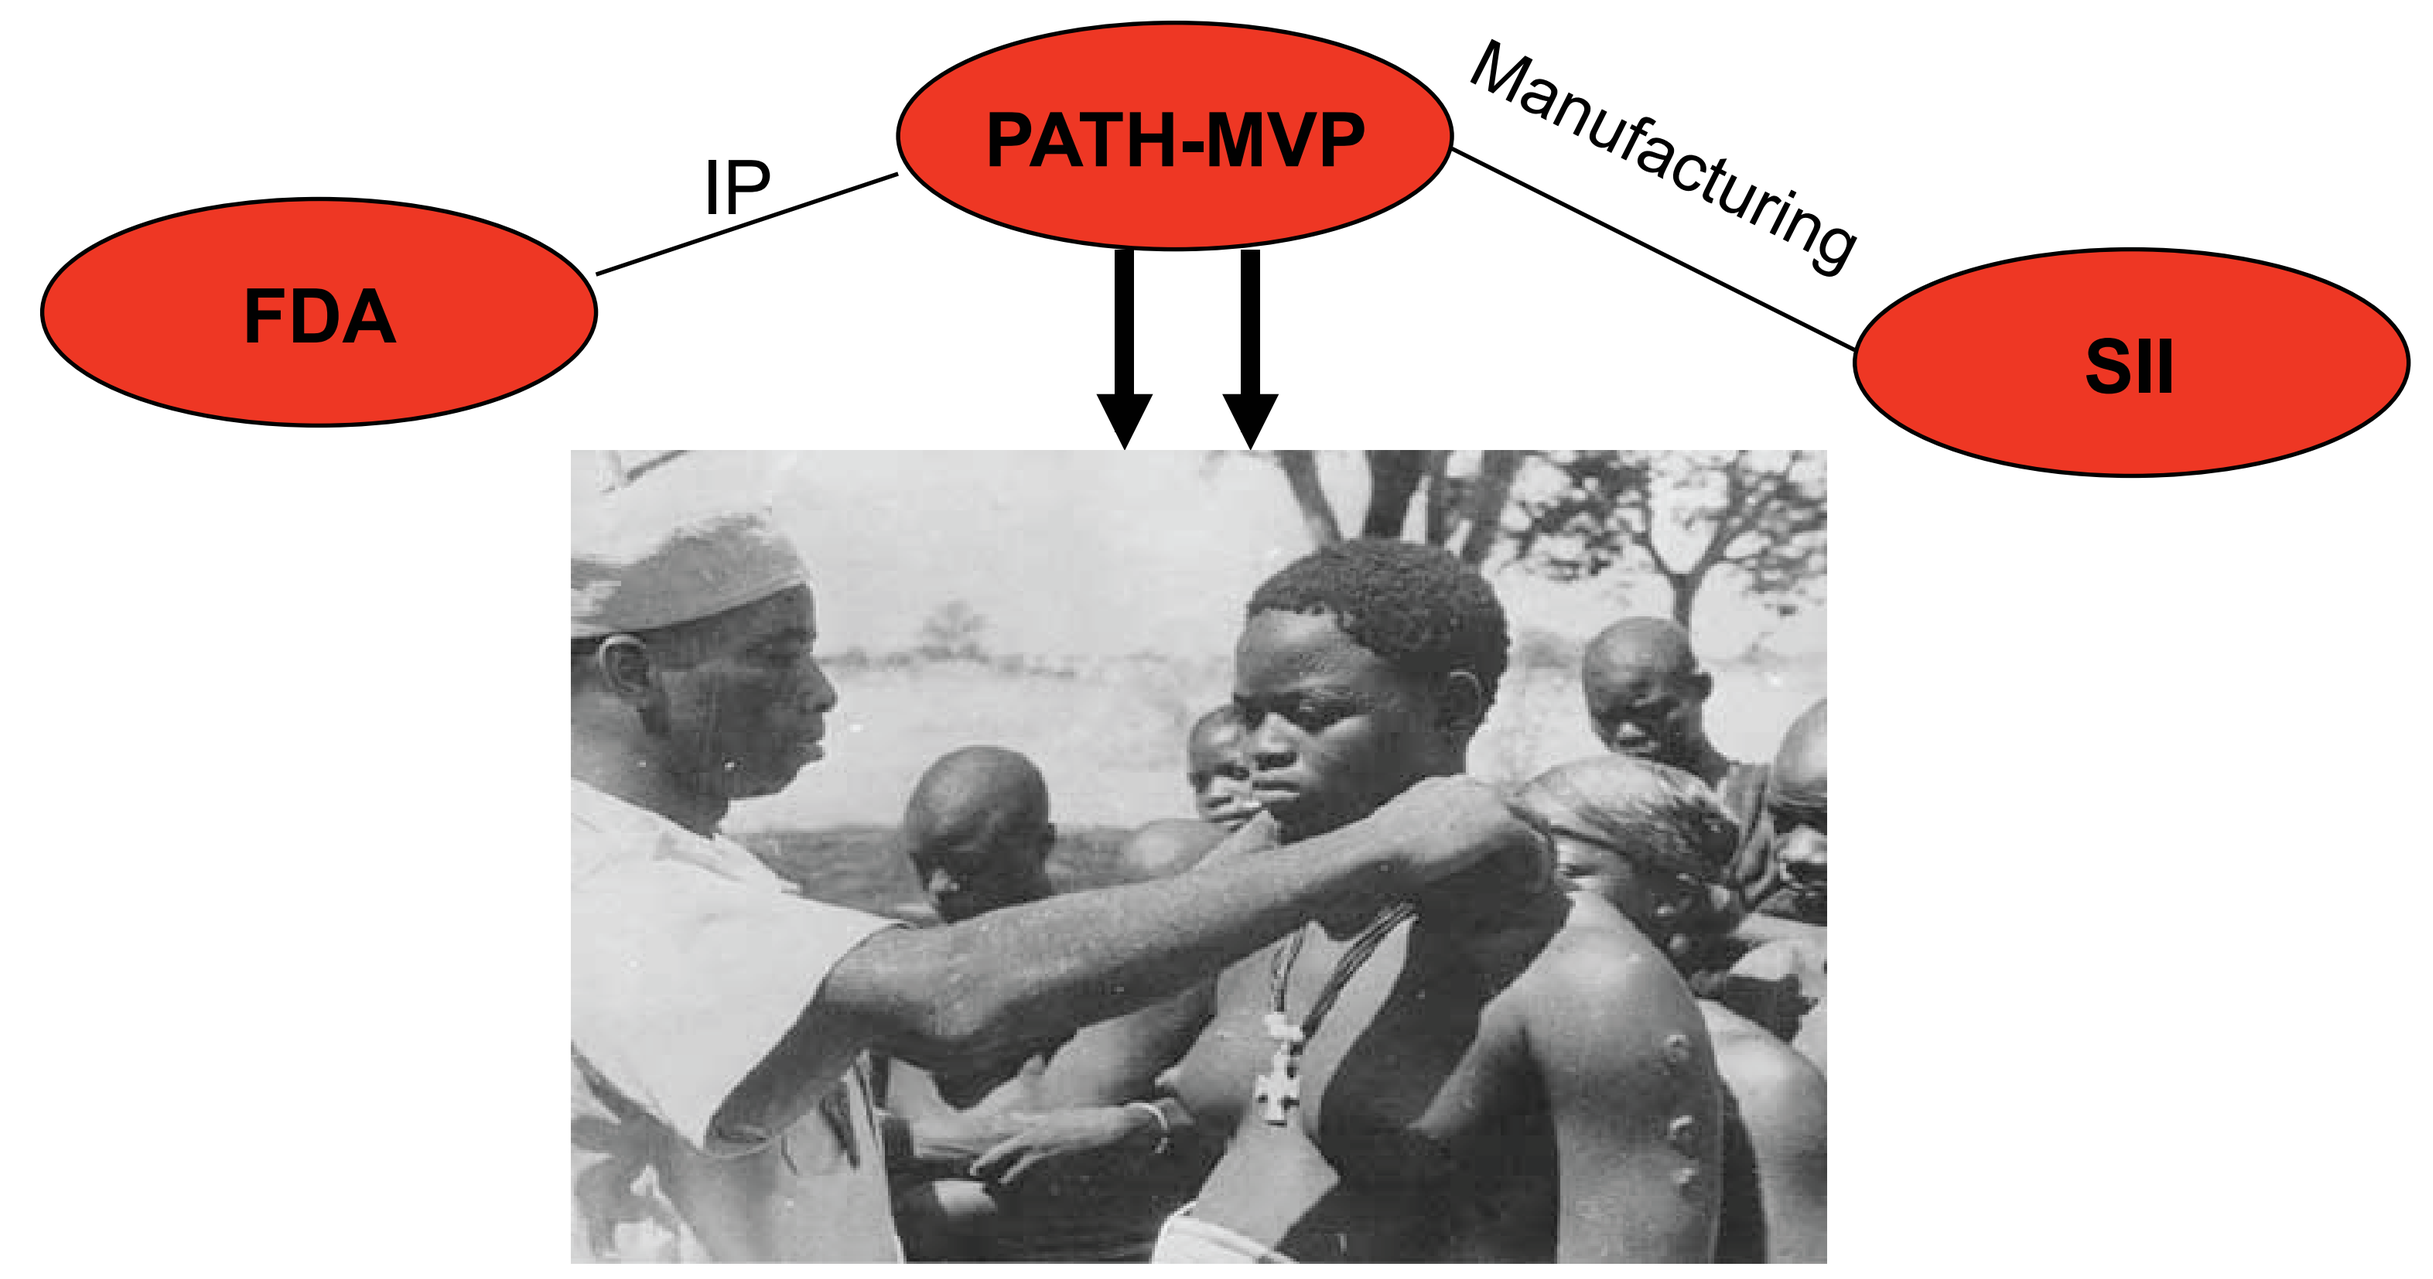

FDA
IP
PATH-MVP
Manufacturing
SII

Role of Partnership Components

<u>FDA</u>
- Provides the IP/technology

<u>PATH</u>
- License Agreement with the FDA
- Funds and manage the project
- Clinical trials

<u>SII (Serum Institute of India)</u>
- Manufactures the vaccine

<u>WHO (World Health Organization)</u>
- Provides expertise regarding epidemiology
- Provides expertise regarding clinical trials

Technology

- Conjugated vaccine
- Efficient conjugation procedure of bacterial polysaccharides to a carrier protein, using dihydrazide.
- IP directed towards Scale-up process.

MVP website: http://www.meningvax.org/

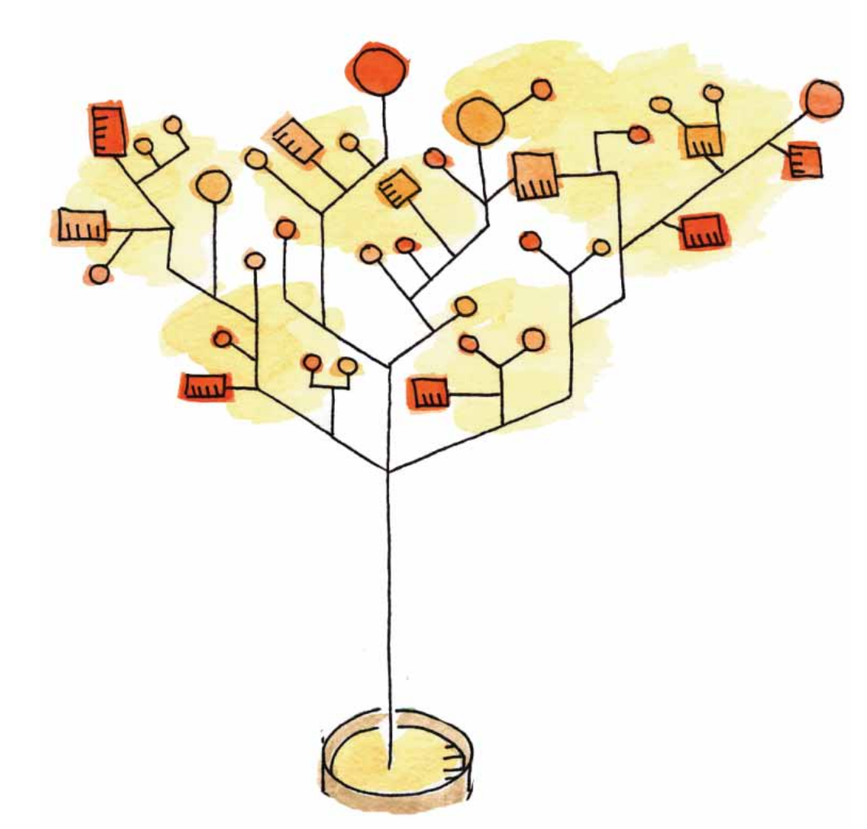